Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® or the author of this poster.

Phase 1/2a trial of nadunolimab, a first-in-class fully humanized monoclonal antibody against IL1RAP, in combination with gemcitabine and nab-paclitaxel (GN) in patients with pancreatic adenocarcinoma (PDAC)

4141

E Van Cutsem[1]; R Eefsen[2]; S Ochsenreither[3,4,5]; Z Zvirbule[6]; A Ivanauskas[7]; D Arnold[8]; E Baltruskeviciene[9]; P Pfeiffer[10]; J Yachnin[11]; G Carbonero[12]; R Greil[13]; C Jungels[14]; L Poulsen[15]; A Awada[14]; I Garcia Ribas[16]; N Losic[16]; J Collignon[17]

[1]UZ Leuven Gasthuisberg, Leuven, Belgium; [2]Herlev og Gentofte Hospital, Herlev, Denmark; [3]Charité Comprehensive Cancer Center, Berlin, Germany; [4]Charité, Department of Hematology, Oncology and Tumor Immunology, Berlin, Germany; [5]German Cancer Consortium (DKTK), Berlin, Germany; [6]Riga East Clinical University Hospital, Riga, Latvia; [7]The Hospital of Lithuanian University of Health Sciences, Kaunas, Lithuania; [8]Asklepios Tumorzentrum Hamburg, AK Altona, Hamburg, Germany; [9]National Cancer Institute, Vilnius, Lithuania; [10]Odense University Hospital, Odense, Denmark; [11]Karolinska University Hospital, Solna, Sweden; [12]Hospital Universitario 12 de Octubre, Madrid, Spain; [13]IIIrd Medical Department, Paracelsus Medical University Salzburg, Salzburg Cancer Research Institute-CCCIT, Cancer Cluster Salzburg, Salzburg, Austria; [14]Institut Jules Bordet, Brussels, Belgium; [15]Aalborg University Hospital, Aalborg, Denmark; [16]Cantargia AB, Lund, Sweden; [17]CHU de Liège, Liège, Belgium

Introduction

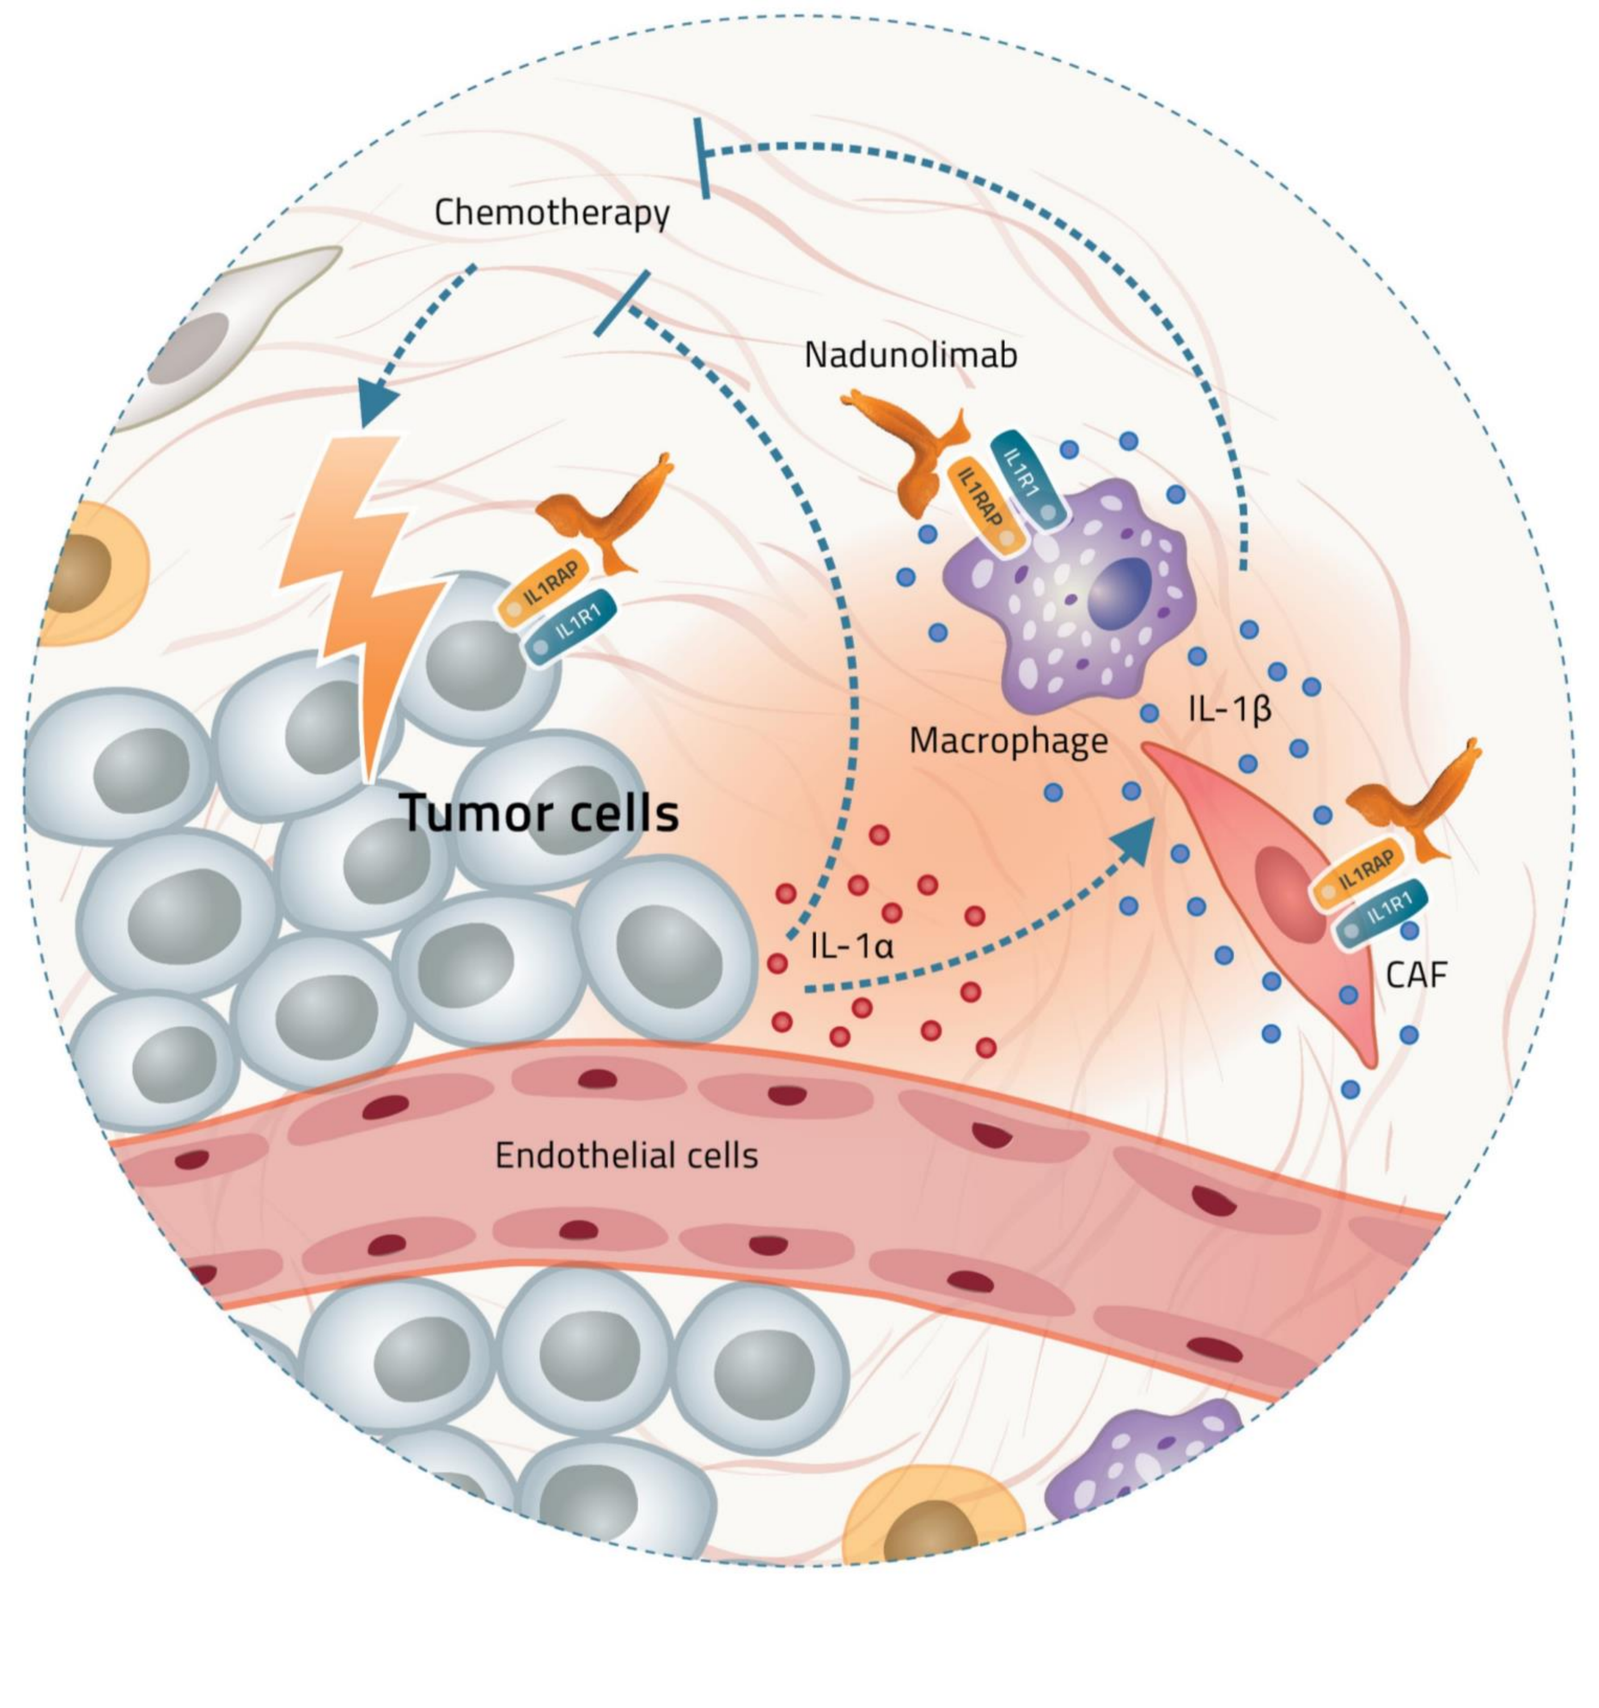
Chemotherapy
Nadunolimab
Macrophage
IL-1β
Tumor cells
IL-1α
CAF
Endothelial cells

Figure 1: Nadunolimab synergizes with chemotherapy.

Interleukin-1 Receptor Accessory Protein (IL1RAP) is expressed on cancer and stromal cells of many solid tumors. IL-1α and IL-1β modulate tumor-promoting and other down-stream factors (e.g. IL-6, IL-8, CRP) by signaling via IL-1 receptor type 1 (IL-1R1), which requires dimerization of IL-1R1 and IL1RAP.

Chemotherapy may trigger IL-1α release by tumor cells, which stimulates release of IL-1β by stromal cells[1-3]. The presence of both forms of IL-1 in the tumor microenvironment contributes to chemoresistance[4-5]. Blockade of both IL-1α and IL-1β may thus constitute an attractive therapeutic approach for cancer in combination with chemotherapy.

Nadunolimab (CAN04) is a fully humanized monoclonal IgG1 antibody targeting IL1RAP. IL1RAP blockade by nadunolimab offers a unique therapeutic approach compared to other IL-1-targeting concepts; it inhibits the tumor-promoting and chemoresistance signals mediated by both IL-1α and IL-1β, and induces ADCC of IL1RAP-expressing cells.

Preliminary interim results of nadunolimab combined with gemcitabine/nab-paclitaxel (GN) in pancreatic ductal adenocarcinoma (PDAC) patients from the ongoing CANFOUR phase I/IIa clinical trial (NCT03267316) have been presented for doses of 5 and 7.5 mg/kg[6]. To evaluate if lower doses of nadunolimab may be more suitable as RP2D, CANFOUR was extended with two new arms to evaluate 2.5 and 1 mg/kg. Here, we report extended preliminary interim results by including also the two lower dose cohorts.

Study Design

Study population (n=76):
- Stage III or IV PDAC
- Chemotherapy naïve
- ECOG PS 0 or 1

Nadunolimab[a] + Gemcitabine/Nab-paclitaxel[c]
5 mg/kg (n=28);
7.5 mg/kg (n=8)
FPI: Mar 2019; LPI: Oct 2020

Nadunolimab[b] + Gemcitabine/Nab-paclitaxel[c]
2.5 mg/kg (n=20);
1 mg/kg (n=20)
FPI: Feb 2021; LPI: Sep 2021

Until PD per iRECIST → **Survival follow-up**

Primary endpoints
Incidence of grade ≥3 AE related to nadunolimab

Secondary endpoints
iORR; iPFS per iRECIST; OS

Exploratory endpoints
- Effects on biomarkers in serum and tumor biopsies

[a]Nadunolimab given Q1W for first 6 wks followed by Q2W; single priming dose (0.5 mg/kg) given on Day -7 to mitigate infusion-related reactions.
[b]Nadunolimab given on Day 1 and 15 in cycles of 28 days and on Day 8 in Cycle 1 only; no priming dose.
[c]Gemcitabine (1000 mg/m^2)/Nab-paclitaxel (125 mg/m^2) given in cycles of 28 days on Day 1, 8 and 15 of each cycle.

Figure 2: Summary of the study design for the PDAC cohorts in part IIa of the CANFOUR study.

Safety population: n=76; efficacy population: modified intention to treat (mITT): n=73. Three patients did not receive chemotherapy due to consent withdrawal (n=2) or clinical deterioration (n=1).

Patient Characteristics

Table 1: Patient demographics and disease characteristics.

	All (n=76)
Age; years	
Median (Range)	62.0 (43-89)
Sex; n (%)	
Female	32 (42%)
Male	44 (58%)
ECOG PS; n (%)	
0	34 (45%)
1	42 (55%)
Stage; n (%)	
III	6 (8%)
IV	70 (92%)
Prior therapies; n (%)	
Adjuvant/neoadjuvant chemotherapy	7 (9%)
Biliary stent	9 (12%)
Radical surgery	9 (12%)

At data-cut-off on 15 April 2022, 16% of patients were on treatment and 59% alive.

Patients were recruited in Belgium (n=23), Denmark (n=14), Germany (n=11), Lithuania (n=10), Latvia (n=8), Sweden (n=4), Spain (n=3), Austria (n=2), and Estonia (n=1).

Results

Efficacy

Table 2: Response parameters.

Efficacy parameter*	mITT (n=73)
iORR [95% CI]	33% [22-45] (24 iPR)
Benefit beyond initial PD	7% (5 patients)
Disease control rate** [95% CI]	73% [61-82]
Median duration of response [95% CI]	6.5 months [5.5-11.1] (range: 3.6-16.5)
iPFS [95% CI]	7.2 months [5.2-8.5] (range: 0.0-18.7)
Median OS [95% CI]	12.7 months [10.0-19.1] (range: 0.1-28.8)
1-year survival	57%

*Efficacy assessed according to iRECIST including response confirmation
**Disease control rate is iPR+iSD+prolonged iUPD (at least two consecutive iUPDs, not followed by any iCPD)

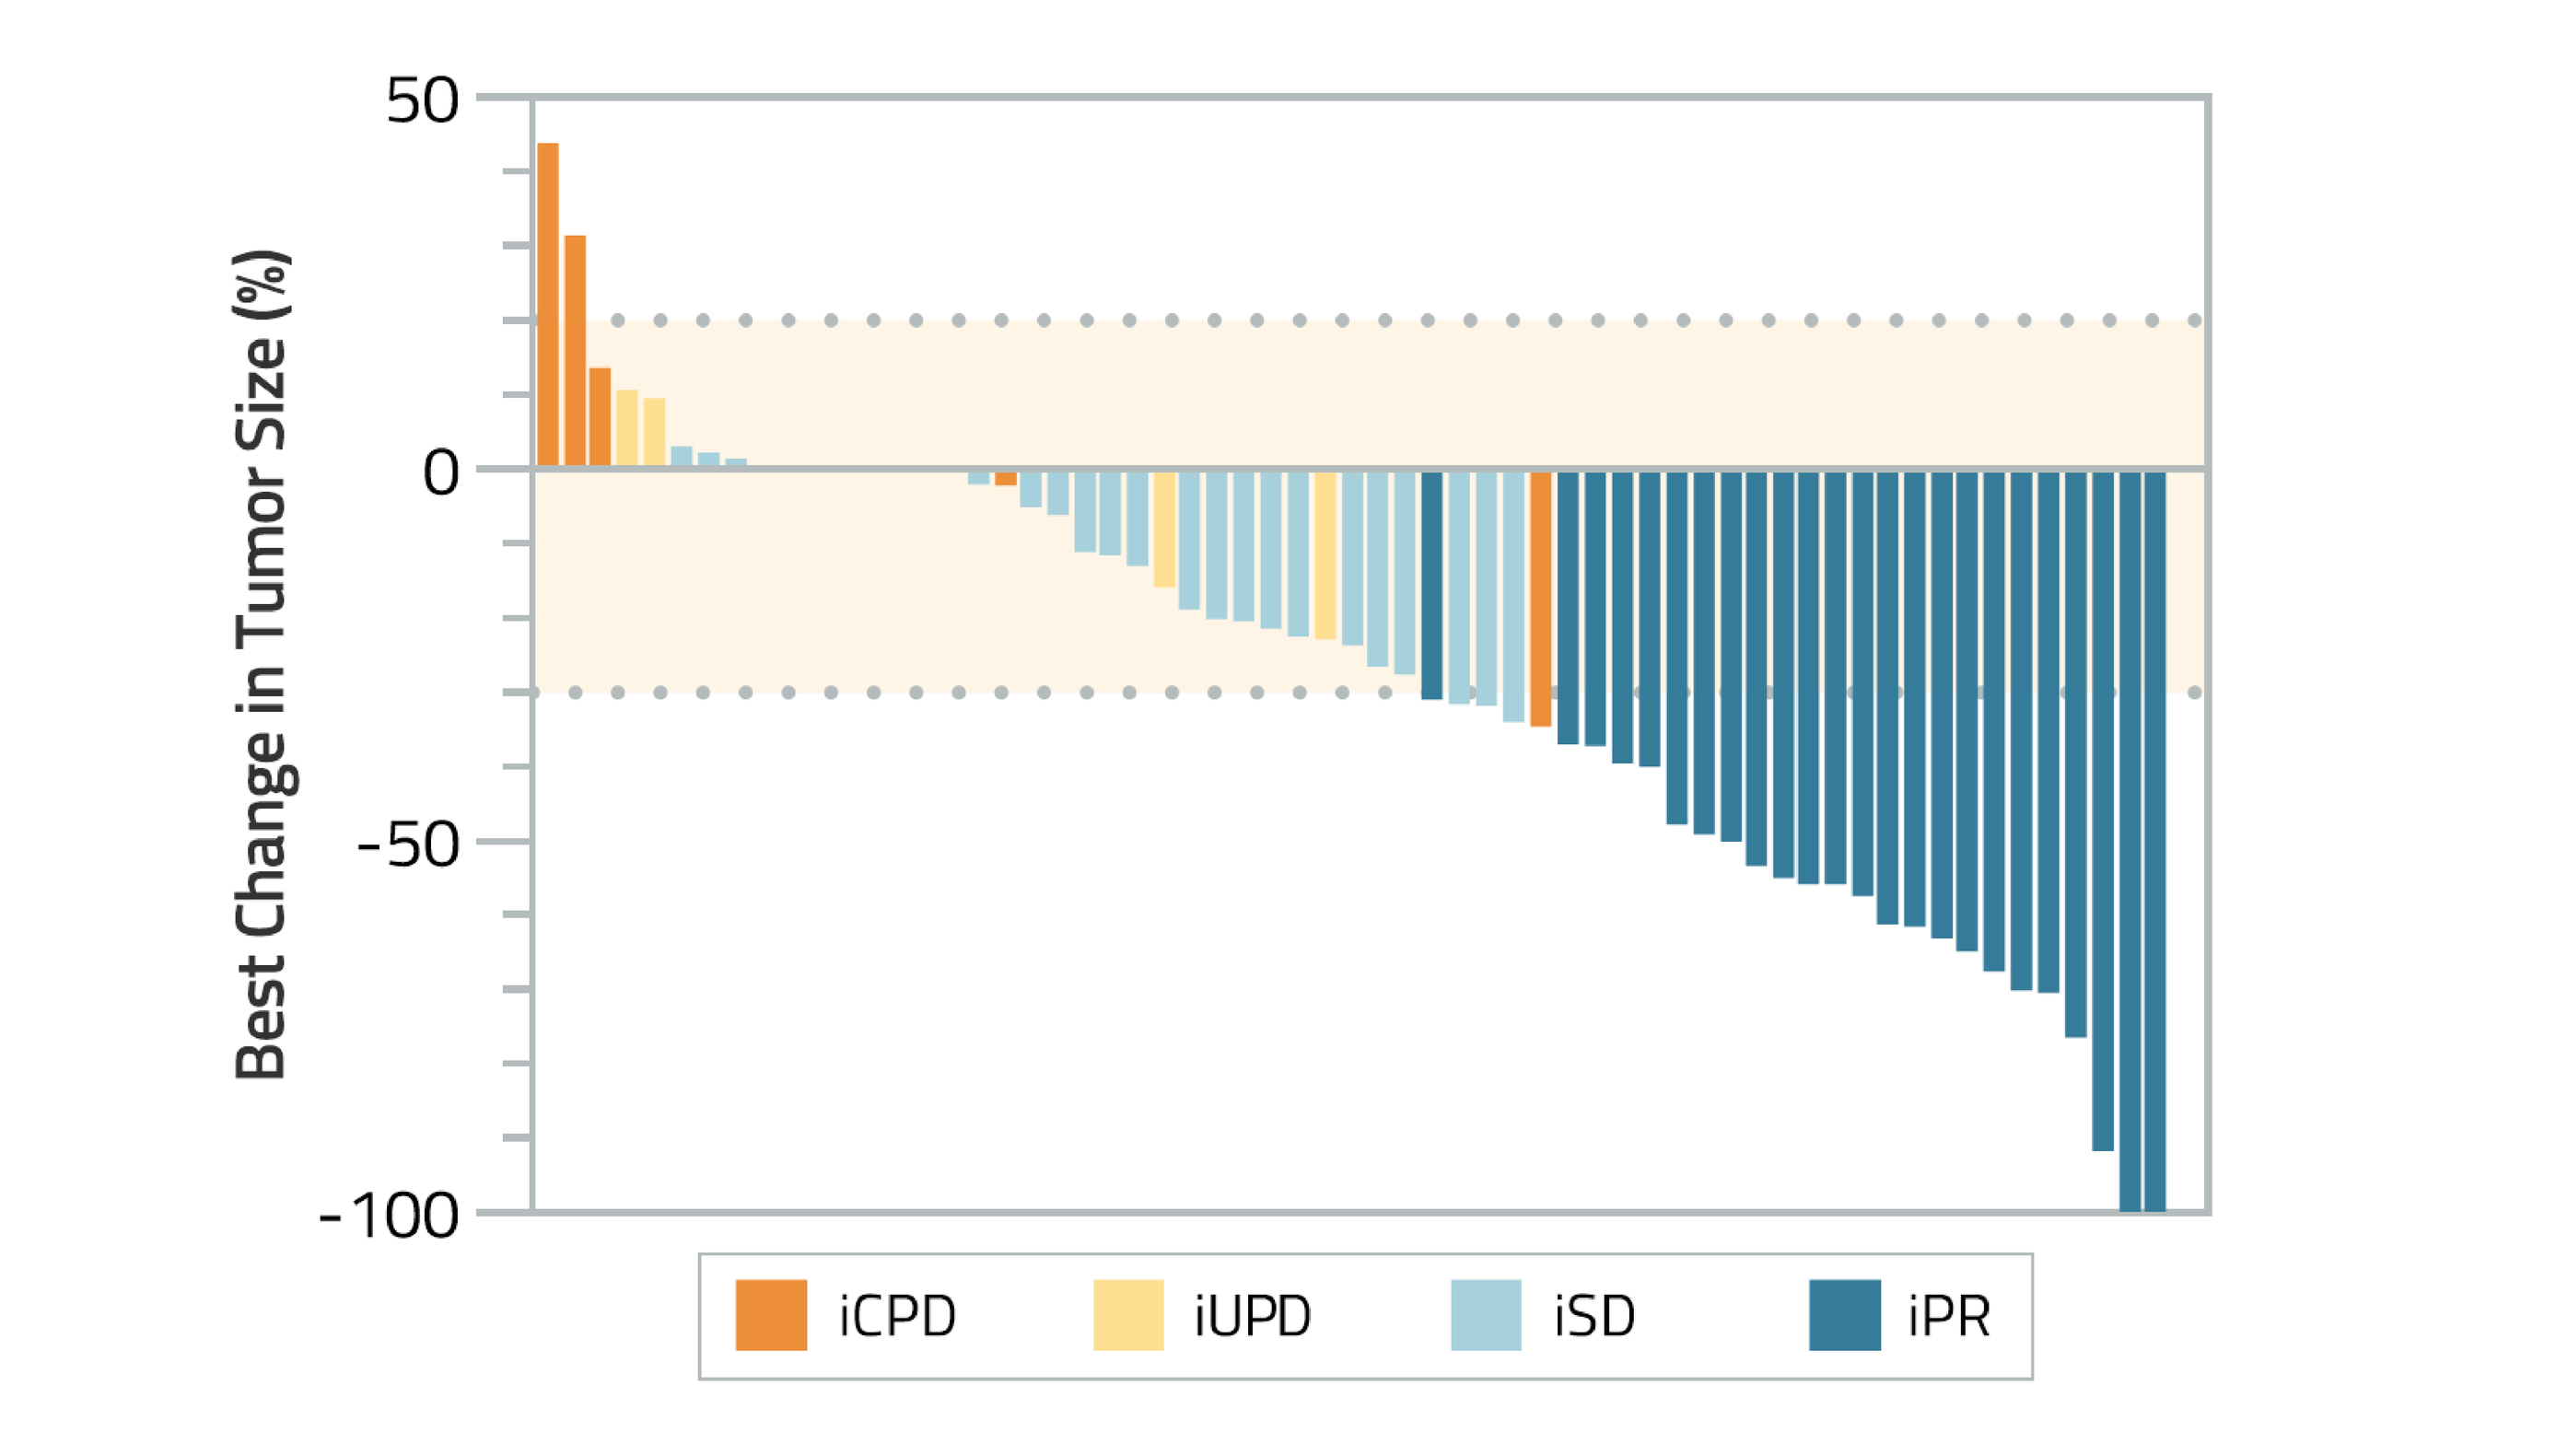
Best Change in Tumor Size (%)
50
0
-50
-100
iCPD
iUPD
iSD
iPR

Figure 3: Tumor response evaluated according to iRECIST. Responses required confirmation.

Twenty-four (33%) patients had iPR as best overall response, 25 (34%) patients had iSD, 8 (11%) patients had iUPD and 5 (7%) patients had iCPD. Five patients with initial PD at first evaluation showed clinical benefit followed by tumor shrinkage, with concomitant CA19-9 reduction, and thus continued therapy.

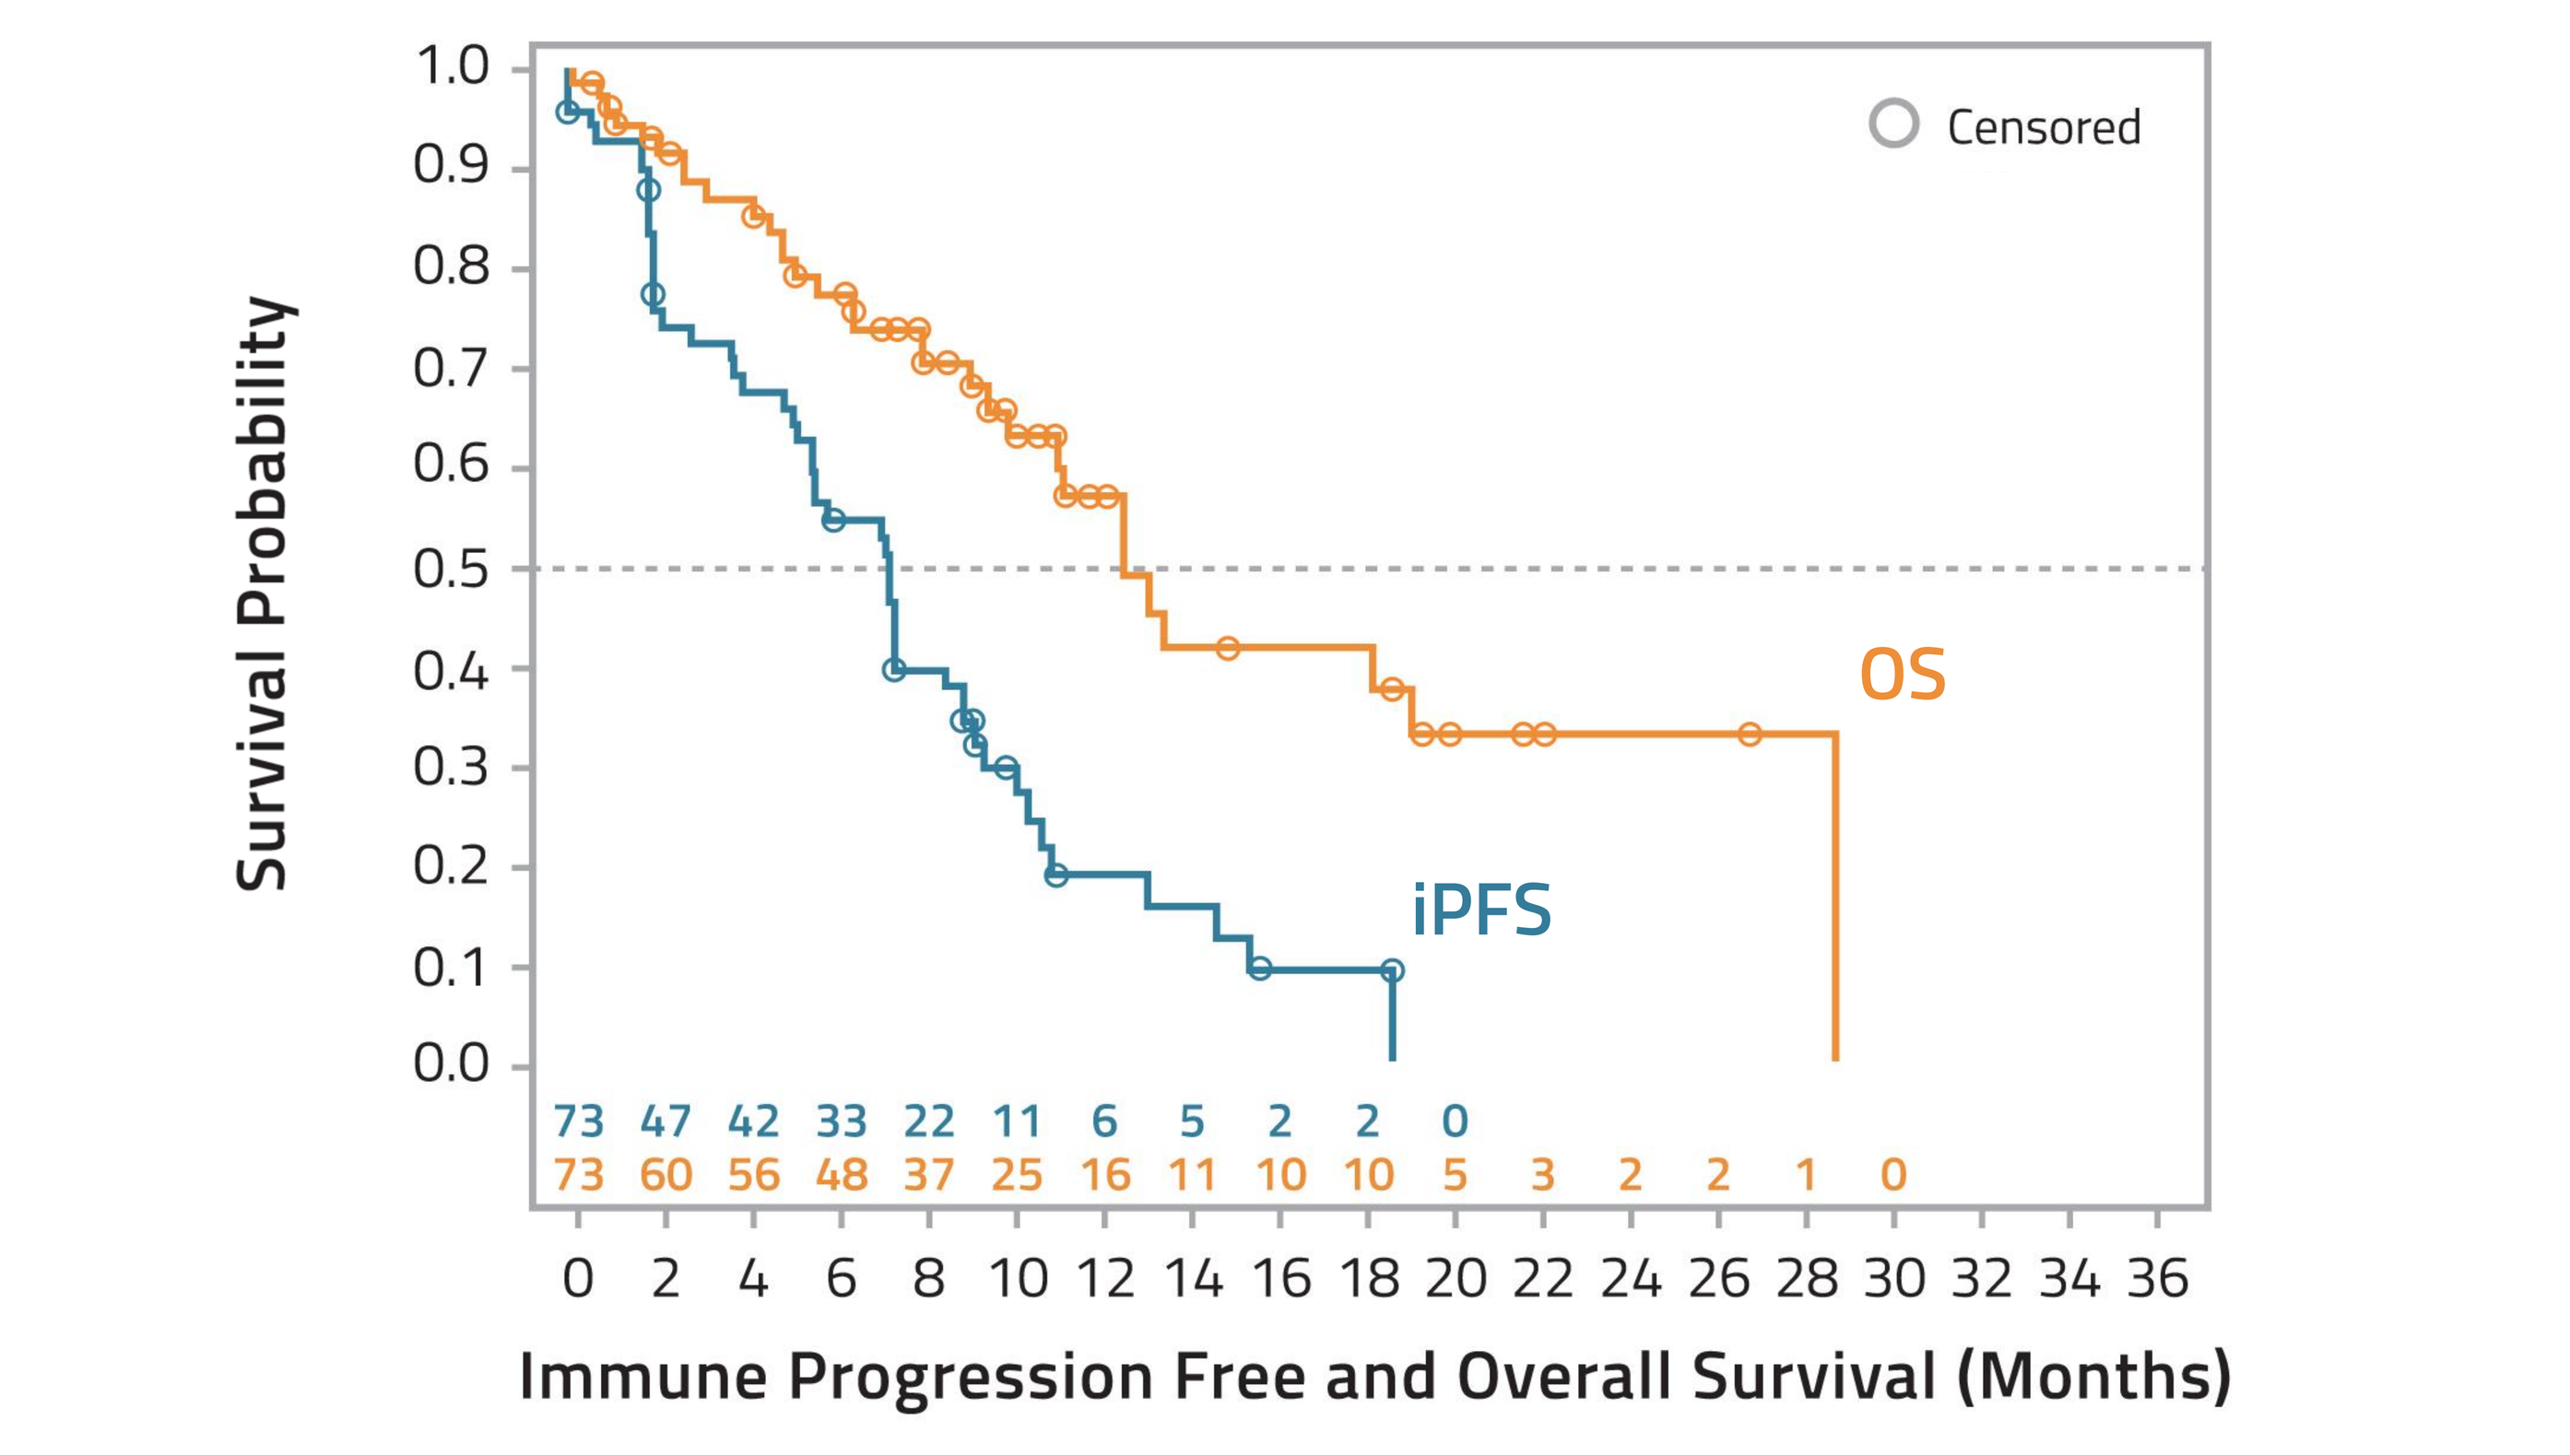
Survival Probability
Censored
OS
iPFS
73 47 42 33 22 11 6 5 2 2 0
73 60 56 48 37 25 16 11 10 10 5 3 2 2 1 0
0 2 4 6 8 10 12 14 16 18 20 22 24 26 28 30 32 34 36
Immune Progression Free and Overall Survival (Months)

Figure 4: OS and iPFS, n=73.

Median OS was 12.7 months (95% CI 10.0-19.1) with events observed in 42%. 6-month survival rate was 77% and 1-year survival rate was 57%. Median iPFS was 7.2 months (95% CI 5.2-8.5) with events observed in 70% of patients. iPFS ranged from 0.0 to 18.7 months. iPFS probability was 55% at 6 months and 19% at 1 year. Twelve patients at data cut-off were still receiving treatment.

Safety

Table 3: Most common TEAE regardless of relationship (worst grade by patient; grades 3-4 in ≥5%).

	Grade 3-4 (n=76)	All grade (n=76)
Hematological TEAE; n (%)		
Neutropenia	49 (65%)	57 (75%)
Leukopenia/WBC decreased	18 (24%)	23 (30%)
Thrombocytopenia	11 (15%)	31 (41%)
Anemia	10 (13%)	37 (49%)
Febrile neutropenia	10 (13%)	10 (13%)
Non-hematological TEAE; n (%)		
GGT increased	13 (17%)	16 (21%)
Hypertension	7 (9%)	10 (13%)
ALT increased	6 (8%)	16 (21%)
Fatigue	6 (8%)	41 (54%)
AST increased	5 (7%)	14 (18%)
Vomiting	5 (7%)	27 (36%)
Cholestasis	4 (5%)	4 (5%)
Hypokalemia	4 (5%)	12 (16%)

Treatment-emergent adverse events (TEAE) of grade 3-4 were reported in 89% of the patients. Only one grade 3 neuropathy was observed.

There were no statistically significant differences in incidences of TEAE across dose levels.

5 mg/kg was considered to be the Maximum Tolerated Dose.

Median duration of nadunolimab treatment was 5.5 months.

Infusion-Related Reactions (IRR):
- Grade 1-2 IRR were observed primarily during the first infusion.
- IRR (all grades) were reported in 29% of patients (grade 3 only in two patients (3%)) and could be managed by standard prophylaxis and ramping infusion.

Neutropenia and Febrile Neutropenia:
- Rates of neutropenia and febrile neutropenia were increased compared to historical controls[7].
- All events of febrile neutropenia and 54% of grade 3-4 neutropenia events occurred during Cycle 1.
- G-CSF given during Cycle 1 was effective in reducing incidence of grade 3-4 neutropenic events. Seven patients were treated prophylactically and only one patient developed grade 3-4 neutropenia (Figure 5).

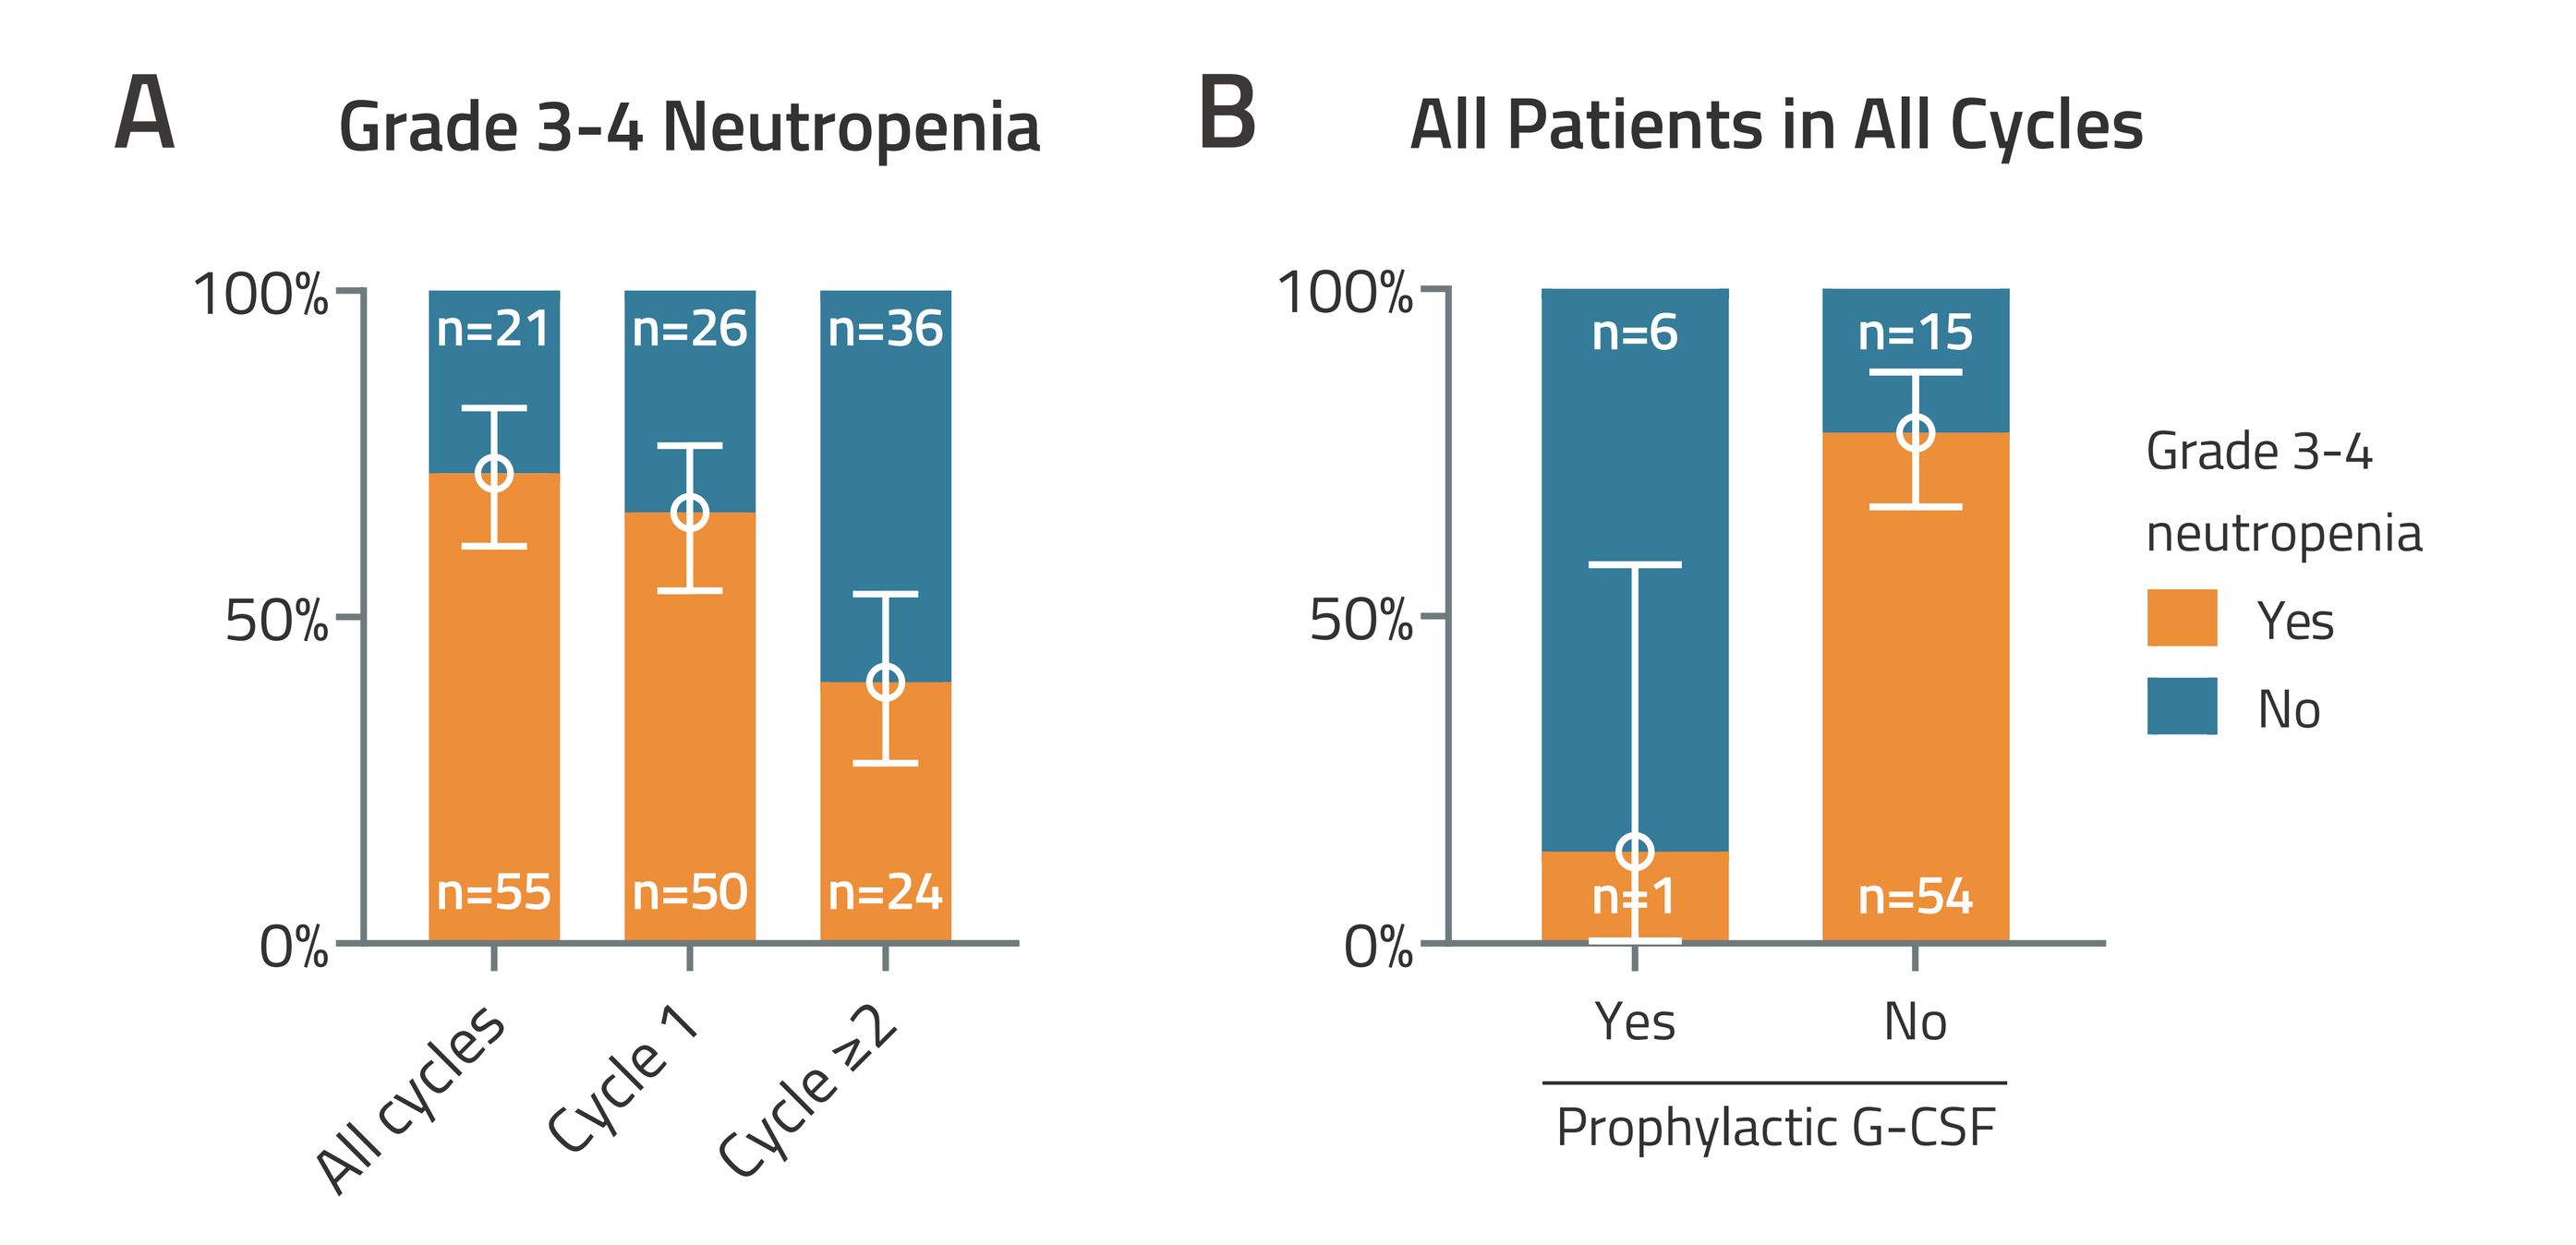
A
Grade 3-4 Neutropenia
100%
50%
0%
n=21
n=26
n=36
n=55
n=50
n=24
All cycles
Cycle 1
Cycle ≥2
B
All Patients in All Cycles
n=6
n=15
n=1
n=54
Yes
No
Prophylactic G-CSF
Grade 3-4 neutropenia
Yes
No

Figure 5: Management of neutropenia. Incidence of grade 3-4 neutropenia (incl. febrile neutropenia) in Cycle 1 and after Cycle 1 (A). Impact of G-CSF primary prophylaxis on prevention of grade 3-4 neutropenia (B).

Biopsy Analyses

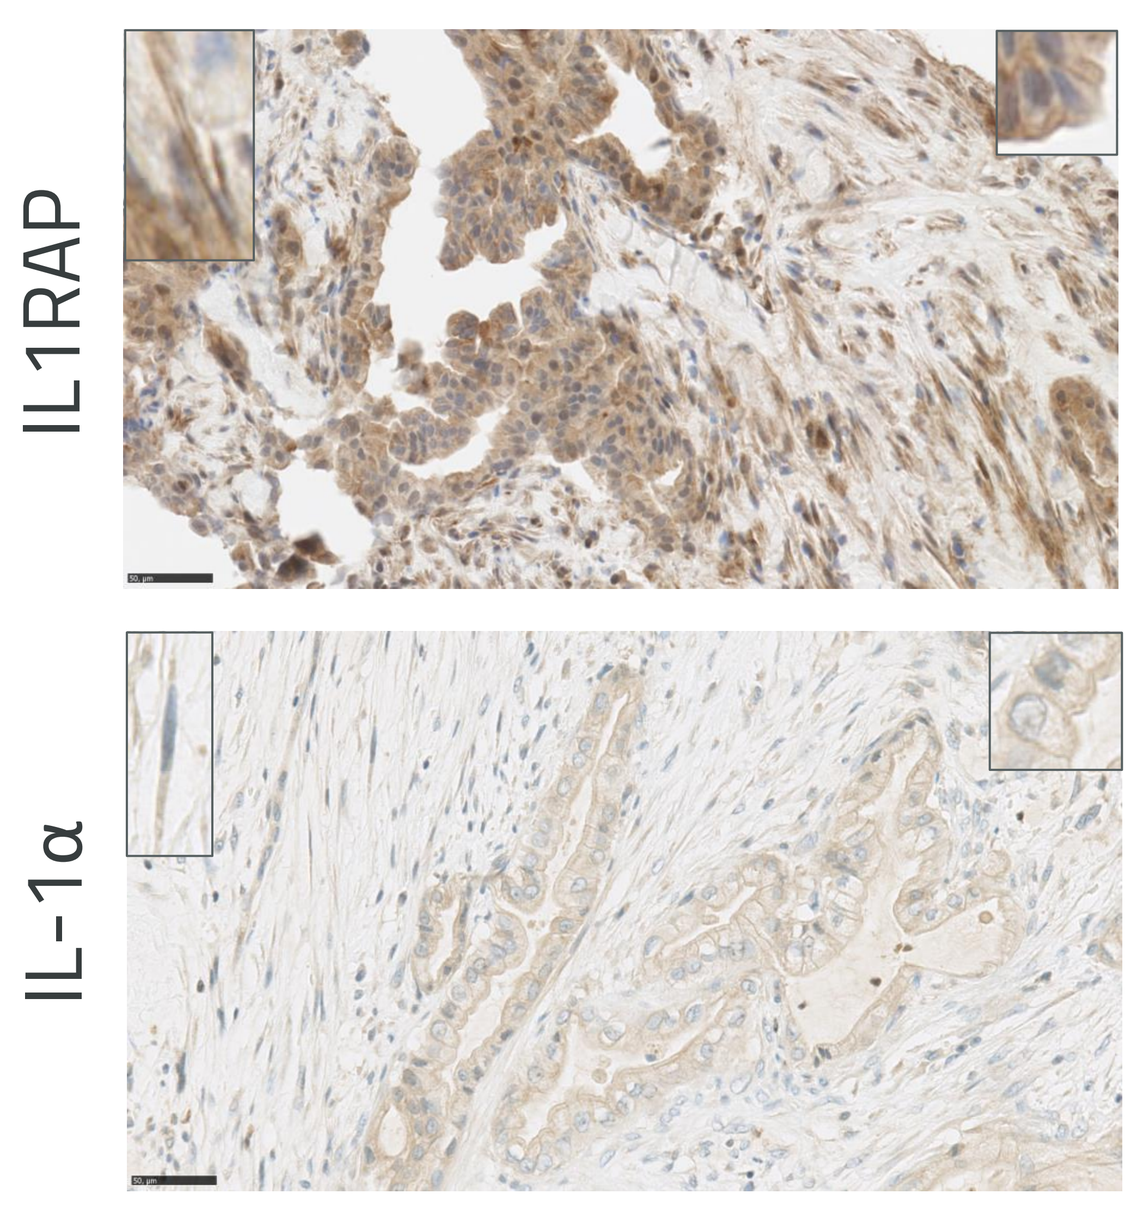
IL1RAP
IL-1α

Figure 6: IL1RAP and IL-1α expression in pre-treatment biopsies.

Screening biopsies were taken at the time of inclusion and stained for IL1RAP and IL-1α expression with immunohistochemistry. Stainings from one representative biopsy are shown.

IL1RAP (top image) and IL-1α (bottom image) were both detected on the surface and intracellularly in tumor cells, cancer-associated fibroblasts and infiltrating immune cells. Positive staining of stromal cells is shown in enlarged inserts in the top left corners, and positive staining of tumor cells in enlarged inserts in the top right corners.

IL-1β was previously shown to be expressed by infiltrating immune cells in PDAC biopsies (data not shown).

Serum Analyses

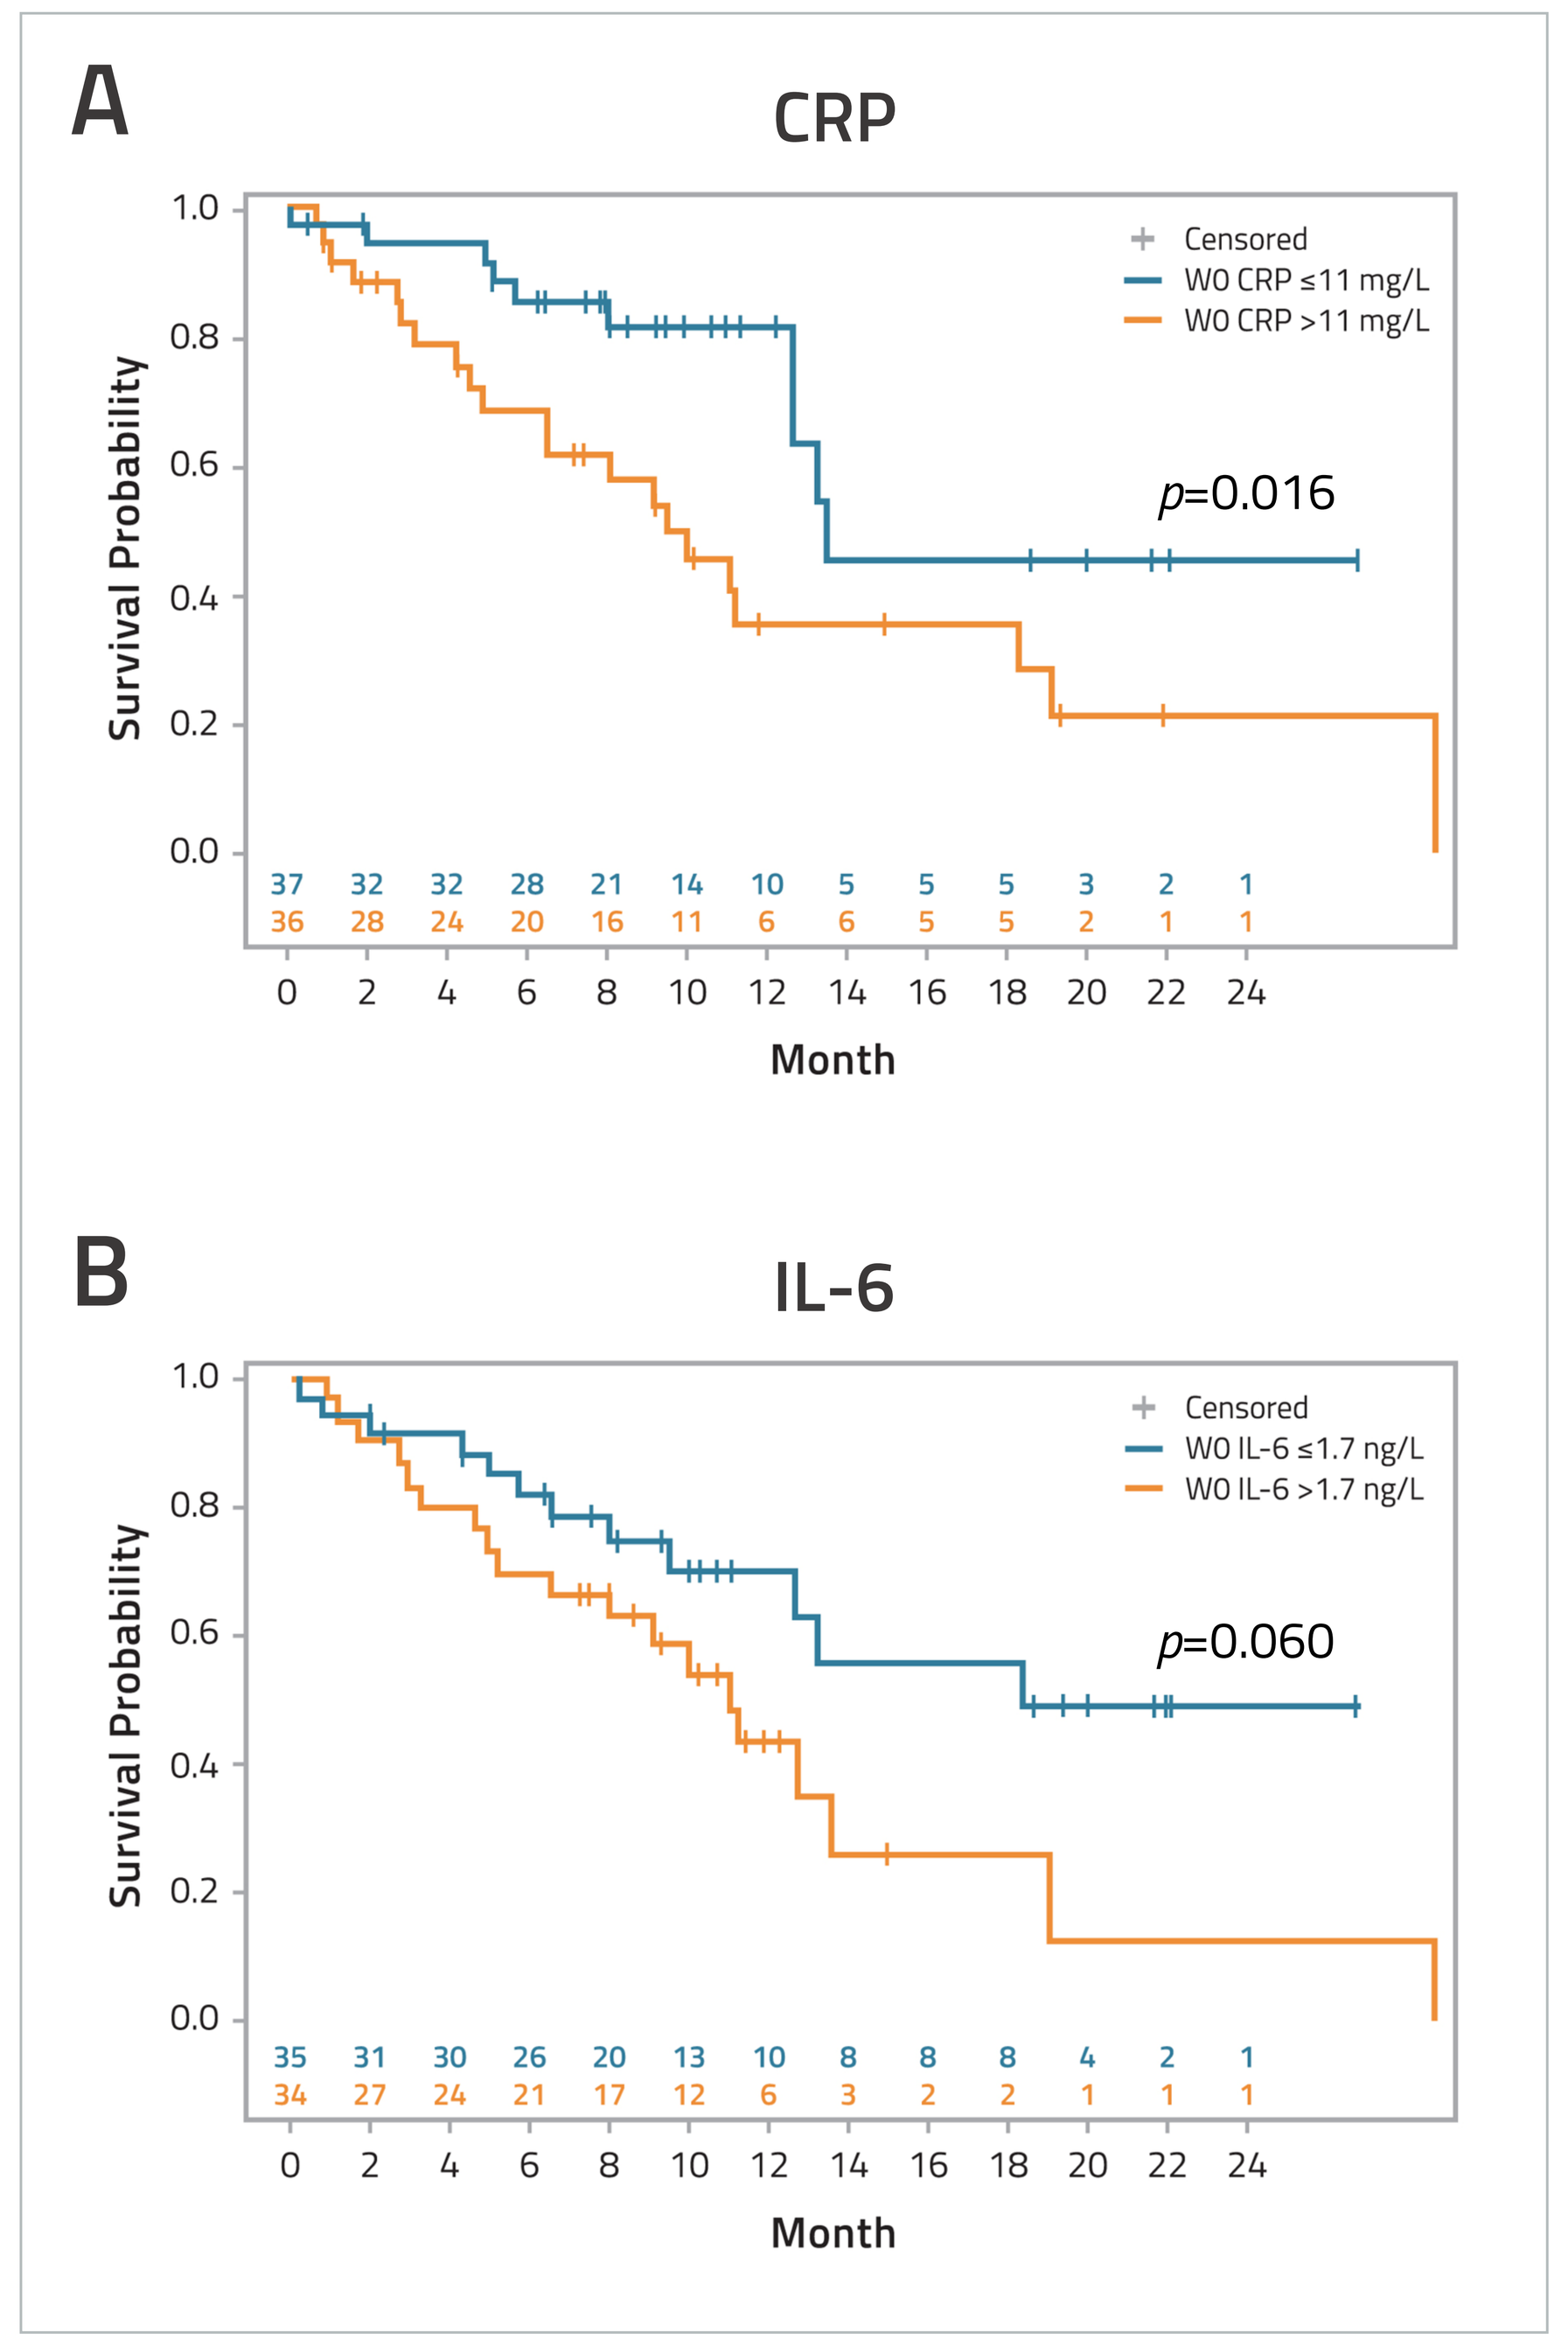
A
CRP
Censored
W0 CRP ≤11 mg/L
W0 CRP >11 mg/L
p=0.016
Survival Probability
37 32 32 28 21 14 10 5 5 5 3 2 1
36 28 24 20 16 11 6 6 5 5 2 1 1
Month
B
IL-6
Censored
W0 IL-6 ≤1.7 ng/L
W0 IL-6 >1.7 ng/L
p=0.060
35 31 30 26 20 13 10 8 8 8 4 2 1
34 27 24 21 17 12 6 3 2 2 1 1 1
Month

Figure 7: Baseline serum levels of CRP and IL-6 correlation with OS.

Serum samples from baseline visit (W0) were analyzed for biomarker levels and correlated against OS. Patients were divided into two groups based on the median value.

CRP correlated with OS and patients with a lower CRP (≤11 mg/l) at baseline had a longer survival as compared to patients with baseline CRP >11 mg/l (13.5 months vs 9.5 months); n=73 (Figure 7A).

Additionally, a lower level of IL-6 (≤1.7 ng/l) at baseline appeared to correlate with longer OS; n=69 (Figure 7B).

Baseline levels of serum IL-8 did not correlate with OS (data not shown).

Conclusions

- Nadunolimab combined with standard gemcitabine/nab-paclitaxel (GN) shows promising efficacy in first-line PDAC:
 - Median iPFS (7.2 months), median OS (12.7 months) and one-year survival (57%) are well above reported values for GN alone[7].
- The overall safety profile is acceptable:
 - Neutropenia during Cycle 1 and grade 1-2 IRR were more common than expected for GN alone[7].
 - All febrile neutropenia and the majority of grade 3-4 neutropenia events occurred in Cycle 1.
 - G-CSF prophylaxis during Cycle 1 reduced the incidence of neutropenia.
 - IRR was managed by standard measures.
 - Notably, grade 3 peripheral neuropathy was lower than historical controls (1% vs 17%)[7].
- Baseline CRP and IL-6 were prognostic for OS.
- A phase II/III trial of nadunolimab and GN in first-line PDAC is in preparation with PanCAN.
- Nadunolimab is currently also evaluated in various indications with chemotherapy or immuno-therapy.

References

- [1]Bruchard et al; Nat Med (2013)
- [2]Chung et al; NPJ Breast Cancer (2022)
- [3]Tjomsland et al; Neoplasia (2011)
- [4]Liu et al; Cancer Res (2018)
- [5]Zhang et al; Cancer Res (2018)
- [6]Awada et al; ESMO Congress (2021)
- [7]Von Hoff et al; N Engl J Med (2013)

Acknowledgements

We would like to thank the patients and their families for participating in the study, and all study staff at the clinical sites. This study was sponsored by Cantargia AB.

cantargia